T1430-05-037

Comparison of the Pharmacokinetics of Four Different Dose-Frequency Combinations of AMZ001 (Diclofenac Gel 3.06%) with Voltaren® (Diclofenac Gel 1%) in Healthy Subjects in a Phase 1 Study

Ahmed A. Kousba[1]; Laetitia Delpy[2]; Dario Carrara[2];
[1]BioAgilytix, San Diego, CA, 92121. [2]Amzell B.V., 2132WT, Hoofddorp, Netherlands.

BioAgilytix

CONTACT INFORMATION:
Ahmed A. Kousba;
ahmed.kousba@bioagilytix.com

PURPOSE

To evaluate how the steady state levels of AMZ001 applied one or two times daily compare to Voltaren® applied four times daily in healthy subjects.

OBJECTIVE(S)

1. **Compare the pharmacokinetic (PK) exposure (C_{max} and AUC_{0-24}) of diclofenac (DCF) following four different dose-frequency combinations of AMZ001 with that of Voltaren®.**
2. **Compare the PK exposure of DCF among the four combinations of AMZ001.**

METHOD(S)

This is a phase 1 study to assess the pharmacokinetics and safety of AMZ001 (Diclofenac [DCF-Na] Gel 3.06%) in comparison with Voltaren® (Diclofenac Gel 1%) in healthy subjects. Seventy-two (72) subjects who met all inclusion and did not meet any exclusion criteria were randomized to parallel groups and received one of following five treatments for 7 consecutive days:

Treatment A. (N = 15 subjects) 32.0 g Voltaren® (320 mg of DCF-Na), applied as 4.0 g per knee four times daily (QID)

Treatment B. (N = 14 subjects) 4.6 g AMZ001 (141 mg of DCF-Na), applied as 1 pump actuation per knee twice daily (BID)

Treatment C. (N = 14 subjects) 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the morning.

Treatment D. (N = 14 subjects) 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the evening.

Treatment E. (N = 15 each) 9.2 g AMZ001 (282 mg of DCF-Na), applied as 2 pump actuations per knee twice daily (BID).

Blood samples were collected on Days 1 and 7 over 24 hours postdose. Two samples were collected on Days 2, 3, 4, 5, and 6; at 7 AM (±1 hour) prior to the first application and at 7 PM (±1 hour) to measure DCF trough level (Ctrough). The plasma was analyzed for DCF by a validated HPLC MS/MS assay.

METHOD(S)(CONTINUED)

DCF PK was calculated using a non-compartmental analysis by validated pharmacokinetic software (WinNonlin Professional v 7.0).

The time to reach a steady state was assessed by linear regression analysis of the Ctrough. To establish the achievement of a steady state, the slope of the linear regression should be equal to the value of zero and the 95% confidence interval of the slope should include zero.

The comparison of the PK exposure of the four different dose-frequency combinations of AMZ001 (Treatments B through E) with Voltaren® (Treatment A) was performed using one-way ANOVA with a p value of significance set to <0.05 using GraphPad Prism v 5.01 (GraphPad Inc., CA). ANOVA was also used for the comparison of the PK exposure among the four AMZ001 treatments.

RESULT(S)

Following topical application of AMZ001 or Voltaren®, DCF was absorbed into the systemic circulation. Day 7 T_{max} was 2-3x shorter than that of Day 1 for the QID and the BID and was similar to that of Day 1 for the QD (Table 1). There was no apparent difference in Days 1 and 7 DCF PK parameters (C_{max}, T_{max} and AUC_{0-24}) following AMZ001 application in the evening and the morning. DCF exposure following 9.2 g AMZ001 gel was ~ 2x higher than that following 4.6 g AMZ001 gel. DCF exposure of Day 7 was higher than that of Day 1 for both AMZ001 and Voltaren® (Tables 1 and 2).

RESULT(S) (CONTINUED)

The steady state of DCF appeared to have been achieved in 3 to 4 days after AMZ001 or Voltaren® application. The mean values for DCF C_{max} and $AUC_{(0-24)}$ on Days 1 and 7 following the AMZ001 in Treatments B, C, D and E were not significantly different from those following Voltaren in Treatment A ($p > 0.05$).

The mean values for DCF Cmax on Days 1 and 7 and for AUC(0-24) on Day 1 following the AMZ001 in Treatments B, D and E were not significantly different from those following AMZ001 in Treatment C ($p > 0.05$). The mean values for DCF $AUC_{(0-24)}$ on Day 7 following the AMZ001 in Treatments B, D and E were significantly different from that of Treatment C. Using Dunnett's multiple comparison post-hoc test showed that, only the mean for $AUC_{(0-24)}$ in Treatment E was significantly different from that of Treatment C.

CONCLUSION(S)

After seven days of treatment, the AUC_{0-24} for Voltaren® Gel was approximately the same as the AUC_{0-24} for AMZ001 administered with 1 pump BID, indicating similar exposure despite the difference in the number of applications (QID versus BID) and lower dose regimens (141 mg vs. 320 mg DCF applied).

aaps PharmSci 360
Advancing Pharmaceutical Sciences, Careers, and Community
ADVANCING PHARMACEUTICAL SCIENCES, CAREERS, AND COMMUNITY

Table 1
Day 1 Plasma PK Parameters of DCF in Healthy Subjects Following Voltaren® in Treatment A and AMZ001 in Treatments B through E

Treatment		T_{max} (h)	C_{max} (ng/mL)	C_{max}/D (ng/mL/mg)	$AUC_{(0-24)}$ (ng*h/mL)	$AUC_{(0-24)}$/D (ng*h/mL/mg)
A	Mean	21.1	5.87	0.0183	61.8	0.193
	SD	3.61	6.77	0.0212	73.1	0.229
B	Mean	21.3	5.73	0.0406	50.7	0.360
	SD	3.65	4.02	0.0285	26.4	0.187
C	Mean	11.9	3.65	0.0259	51.2	0.363
	SD	6.63	2.06	0.0146	35.1	0.249
D	Mean	11.9	4.10	0.0291	47.2	0.335
	SD	5.46	4.01	0.0285	41.9	0.297
E	Mean	20	8.07	0.0286	76.7	0.272
	SD	4.47	8.99	0.0319	62.6	0.222

Treatment A. 32.0 g Voltaren® (320 mg of DCF-Na), applied as 4.0 g per knee four times daily (QID). Treatment B. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 1 pump actuation per knee twice daily (BID). Treatment C. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the morning. Treatment D. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the evening. Treatment E. 9.2 g AMZ001 (282 mg of DCF-Na), applied as 2 pump actuations per knee twice daily (BID)

Table 2
Day 7 Plasma PK Parameters of DCF in Healthy Subjects Following Voltaren® in Treatment A and AMZ001 in Treatments B through E

Treatment		T_{max} (h)	C_{max} (ng/mL)	C_{max}/D (ng/mL/mg)	$AUC_{(0-24)}$ (ng*h/mL)	$AUC_{(0-24)}$/D (ng*h/mL/mg)	$AUC_{(0-tau)}$ (ng*h/mL)	$AUC_{(0-tau)}$/D (ng*h/mL/mg)	C_{min} (ng/mL)	T_{min} (h)	C_{avg} (ng/mL)
A	Mean	1.47	16.9	0.0527	252	0.789	68.6	0.215	7.70	4.27	11.4
	SD	2.56	11.3	0.0354	143	0.448	45.7	0.143	5.53	1.98	7.62
B	Mean	5.57	17.6	0.125	253	1.80	130	0.920	6.26	7.43	10.8
	SD	4.16	10.2	0.0724	128	0.906	67.3	0.478	3.71	4.67	5.61
C	Mean	9.29	12.2	0.0864	169	1.20	169	1.20	4.08	8.86	7.06
	SD	5.69	7.82	0.0554	113	0.804	113	0.804	3.36	9.37	4.72
D	Mean	9.43	15.0	0.106	169	1.20	169	1.20	2.25	11.3	7.04
	SD	4.93	12.6	0.0893	131	0.927	131	0.927	1.17	11.2	5.45
E	Mean	3.20	21.0	0.0746	364	1.29	164	0.582	7.74	8.67	13.7
	SD	3.61	25.6	0.0908	352	1.25	166	0.590	6.68	4.32	13.9

Treatment A. 32.0 g Voltaren® (320 mg of DCF-Na), applied as 4.0 g per knee four times daily (QID). Treatment B. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 1 pump actuation per knee twice daily (BID). Treatment C. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the morning. Treatment D. 4.6 g AMZ001 (141 mg of DCF-Na), applied as 2 pump actuations per knee once daily (QD) in the evening. Treatment E. 9.2 g AMZ001 (282 mg of DCF-Na), applied as 2 pump actuations per knee twice daily (BID)