#T2368

Toxicokinetics of CBX129801, a Bio-active C-Peptide for Potential Replacement Therapy in Type 1 Diabetic Peripheral Neuropathy, Following Subcutaneous Injection of CBX129801 in Monkeys in a 9-Month Study with a 7-Week Recovery Period

BioAgilytix

A. Kousba[1], M. Mazzoni[2], D. Naas[3];

[1]BioAgilytix, San Diego, CA, 92121. [2]Cebix Incorporated, La Jolla, CA, 92037. [3]ProDev Consulting Services, Ltd., Poway, CA, 92064.

ABSTRACT

Purpose:

To assess the toxicokinetics (TK) of CBX129801 in Cynomolgus monkeys following once weekly subcutaneous injection for 39 doses.

Methods:

Four groups received subcutaneous injections of CBX129801 at 0, 0.4, 1.33 and 4.0 mg/kg/week for 39 doses. Blood samples were collected on Days 0, 84, 175 and 266 and during the recovery period at predetermined time. Additional samples were collected during the recovery period for 1.33 and 4.0 mg/kg/week doses on Days 196, 210, 224 (6-month recovery) and 287, 301 and 315 (9-month recovery). The plasma was assayed for CBX129801 by a validated ELISA assay. TK parameters were determined by modelindependent methods.

Results:

Following the first subcutaneous injection of CBX129801 at 0.4, 1.33 and 4.0 mg/kg, mean C_{max} values were 47.6, 233 and 721 nM in males and 67.7, 222 and 750 nM in females, respectively.

Corresponding mean AUCtau values were 215, 1060 and 3270 nM·day in males and 308, 1070 and 3410 nM·day in females. Apparent T1/2 mean values were similar between gender and dose groups and ranged from 2.44 to 3.75 days.

CBX129801 exposure increased as a function of doses in a dose-proportional manner and was similar between the genders. CBX129801 appeared to achieve a steady state within 28 days and maintained high concentrations through a week after the last dose. The plasma concentrations of CBX129801 markedly decreased over time and were substantially low by the end of the recovery period.

Conclusions:

Upon subcutaneous administration to monkeys, CBX129801 exposure was dose proportional, had a linear TK and no gender differences. CBX129801 exposure did not increase with repeated weekly administration of 39 doses.

Purpose

To assess the toxicokinetics (TK) of CBX129801 in Cynomolgus monkeys following once weekly subcutaneous injection for 39 doses.

Introduction

Type 1 diabetes is characterized by the body's inability to produce proinsulin and consequently both insulin and C-peptide.

It is estimated that 4 million people in the U.S. and Europe have type 1 diabetes, and about 15,000 children are diagnosed with type 1 diabetes in the U.S. each year.

Scientific data suggests that C-peptide deficiency in type 1 is a contributing cause of many of the long-term complications associated with type 1 diabetes, despite insulin replacement therapy.

A protective effect of C-peptide treatment was demonstrated in preclinical and early clinical studies.

Cebix is developing a long-acting C-peptide drug (CBX129801; PEGylated C-peptide) for the treatment of long-term complications of diabetes.

This work presents the TK of CBX129801 in monkeys.

Methods

Study Design

Four groups of monkeys received subcutaneous injections of CBX129801 at 0, 0.4, 1.33 and 4.0 mg/kg/week for 39 doses. Blood samples were collected on Days 0, 84, 175 and 266 and during the recovery period at predetermined time. Additional samples were collected during the recovery period for 1.33 and 4.0 mg/kg/week doses on Days 196, 210, 224 (6-month recovery) and 287, 301 and 315 (9-month recovery).

Analytical Method

The plasma was assayed for CBX129801 by a validated ELISA assay.

TK Analysis

The TK parameters were determined by standard modelindependent methods (Gibaldi and Perrier, 1982) using Phoenix WinNonlin Professional Version 6.1 (Pharsight Corp., Mountain View, CA).

Figure 1

Mean Plasma Concentrations of CBX129801 in Monkeys Following Subcutaneous Injections of CBX129801 in a 3/6/9-Month Toxicity Study

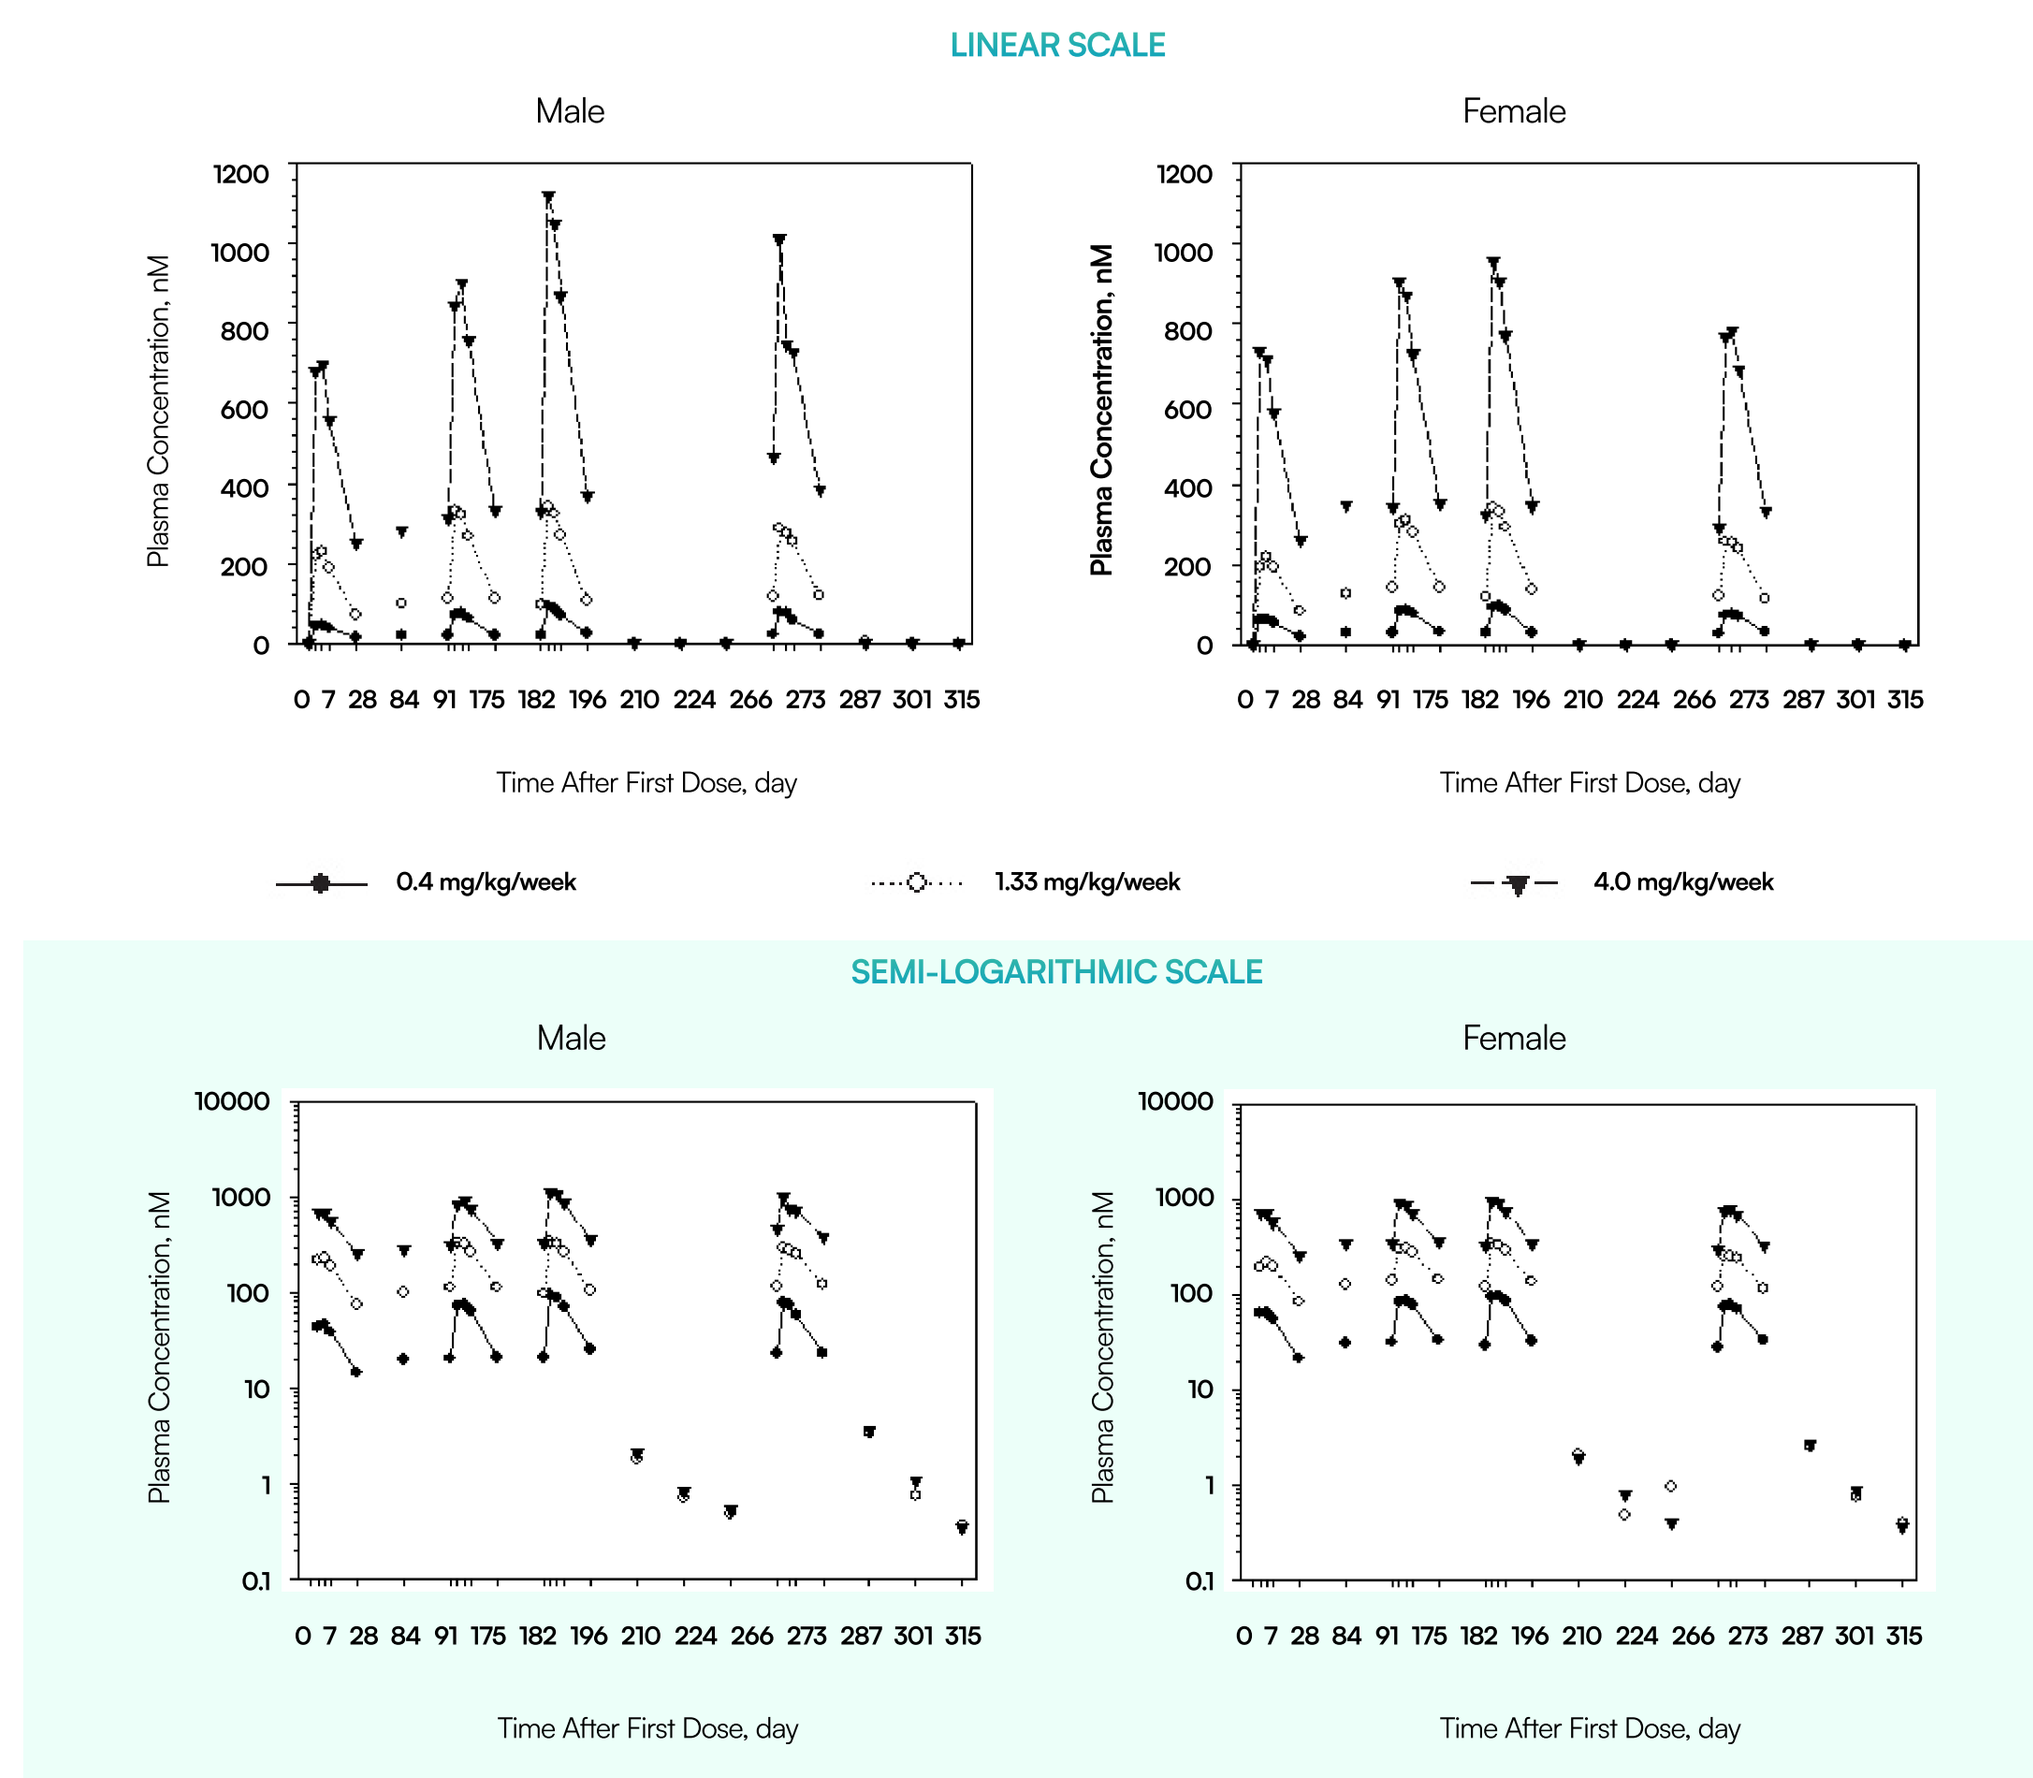

LINEAR SCALE
Male
Female
Plasma Concentration, nM
Time After First Dose, day
0.4 mg/kg/week
1.33 mg/kg/week
4.0 mg/kg/week
SEMI-LOGARITHMIC SCALE

Figure 2

CBX129801 C_{max} versus CBX129801 Dose in Rats Following Subcutaneous Injections of CBX129801 in a 3/6-Month Toxicity Study

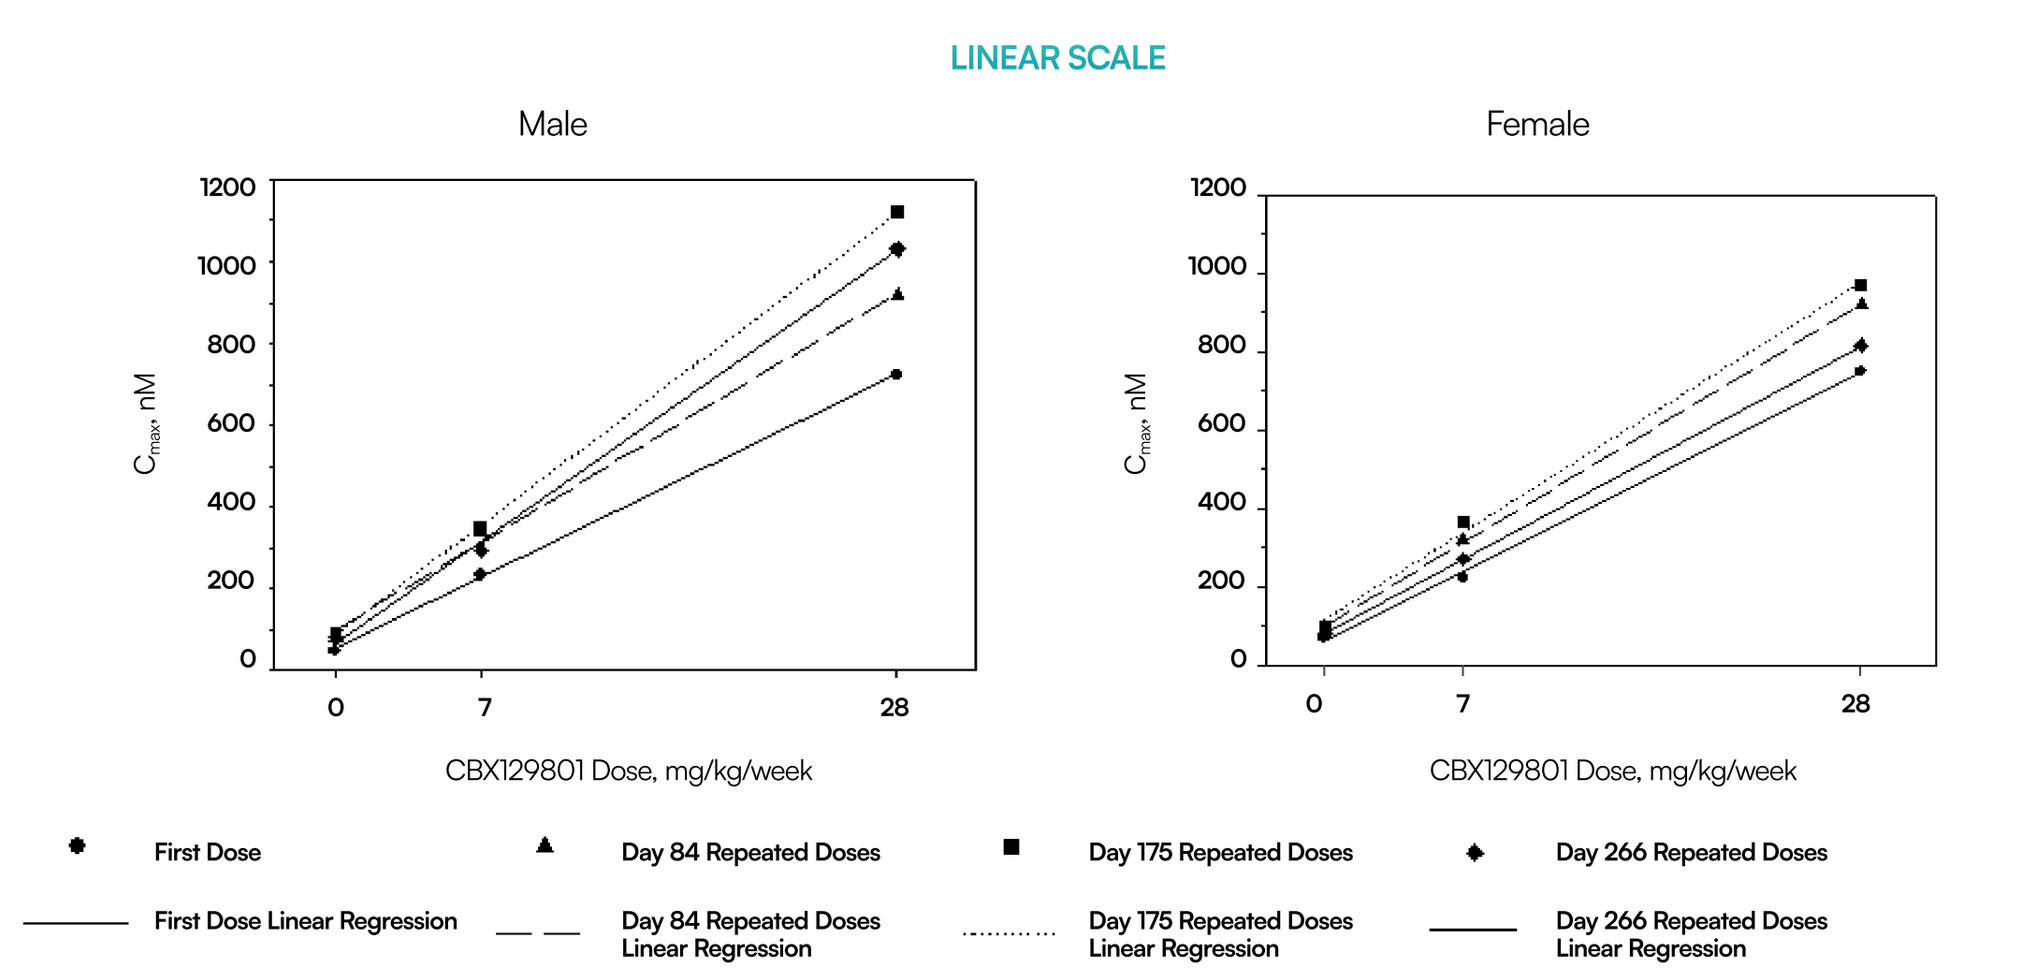

LINEAR SCALE
Male
Female
CBX129801 Dose, mg/kg/week
First Dose
Day 84 Repeated Doses
Day 175 Repeated Doses
Day 266 Repeated Doses
First Dose Linear Regression
Day 84 Repeated Doses Linear Regression
Day 175 Repeated Doses Linear Regression
Day 266 Repeated Doses Linear Regression

Figure 3

CBX129801 Dose-Normalized Predose Plasma Concentrations (nM/mg) in Monkeys Following Subcutaneous Injections of CBX129801 in a 3/6/9-Month Toxicity Study

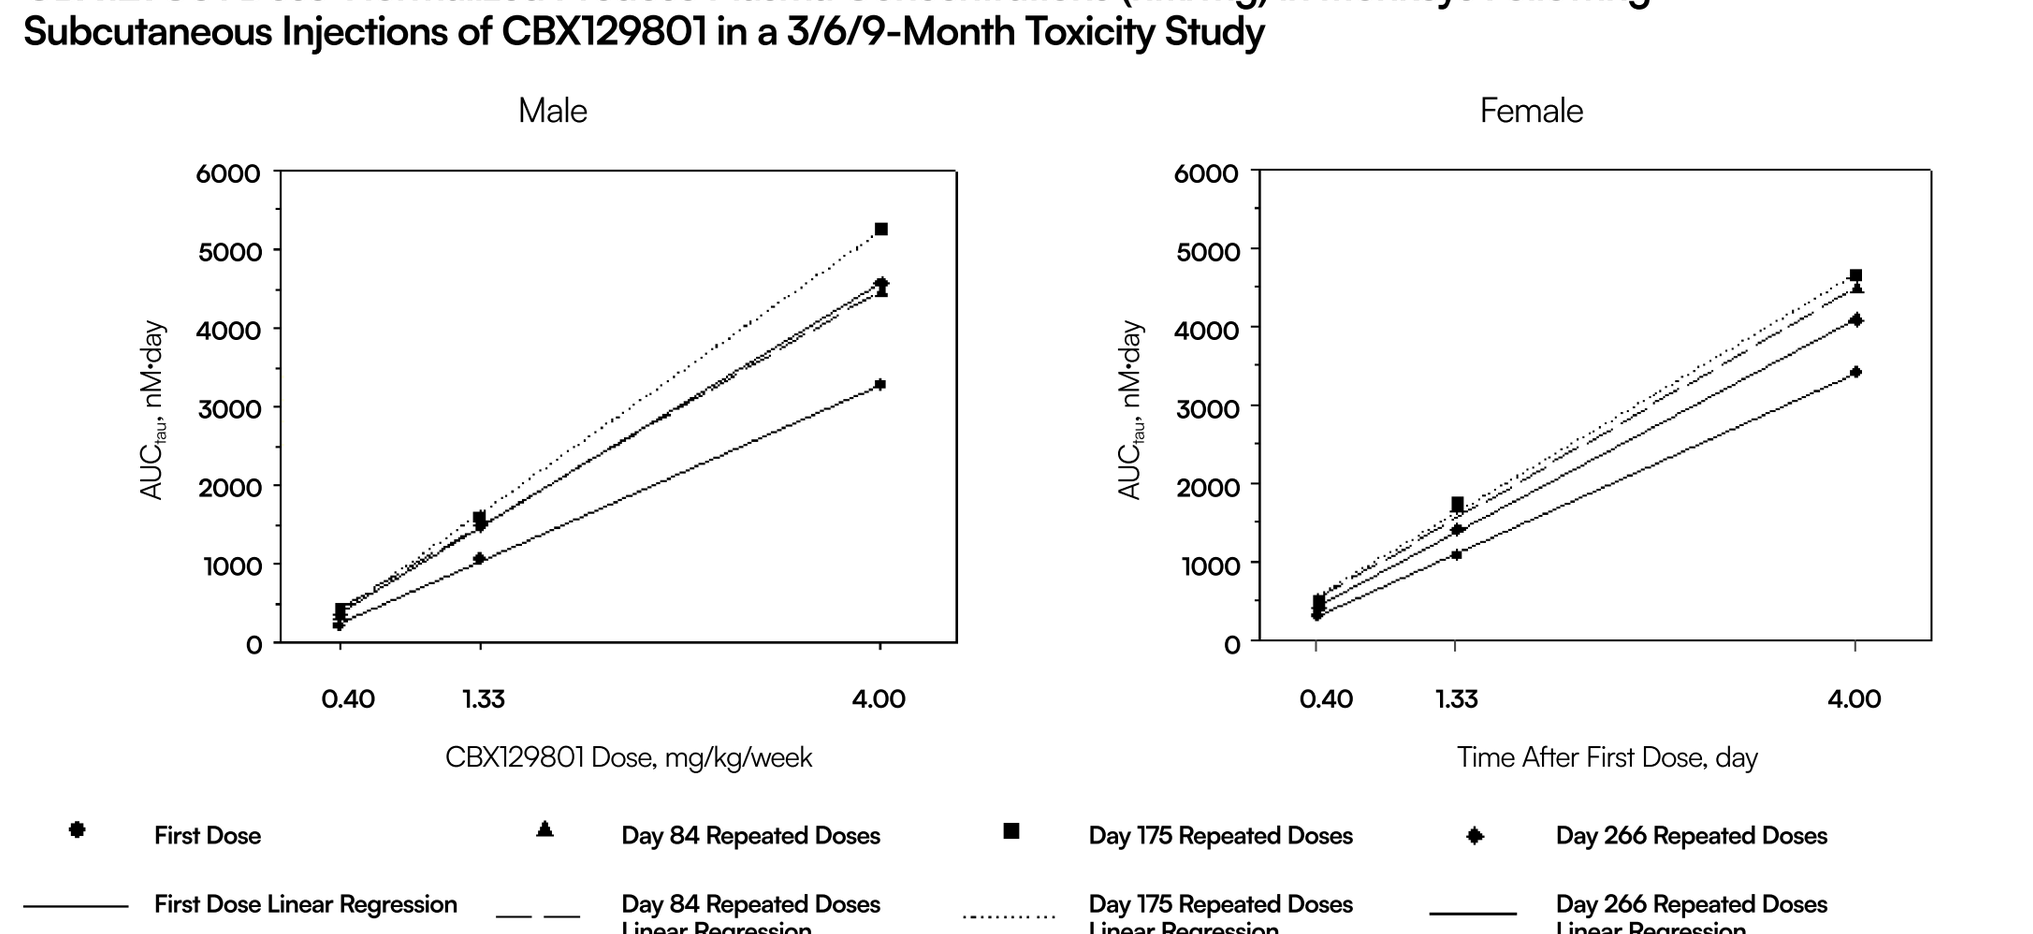

Male
Female
CBX129801 Dose, mg/kg/week
Time After First Dose, day
First Dose
Day 84 Repeated Doses
Day 175 Repeated Doses
Day 266 Repeated Doses
First Dose Linear Regression
Day 84 Repeated Doses Linear Regression
Day 175 Repeated Doses Linear Regression
Day 266 Repeated Doses Linear Regression

Figure 4

CBX129801 C_{max} versus CBX129801 Dose in Rats Following Subcutaneous Injections of CBX129801 in a 3/6-Month Toxicity Study

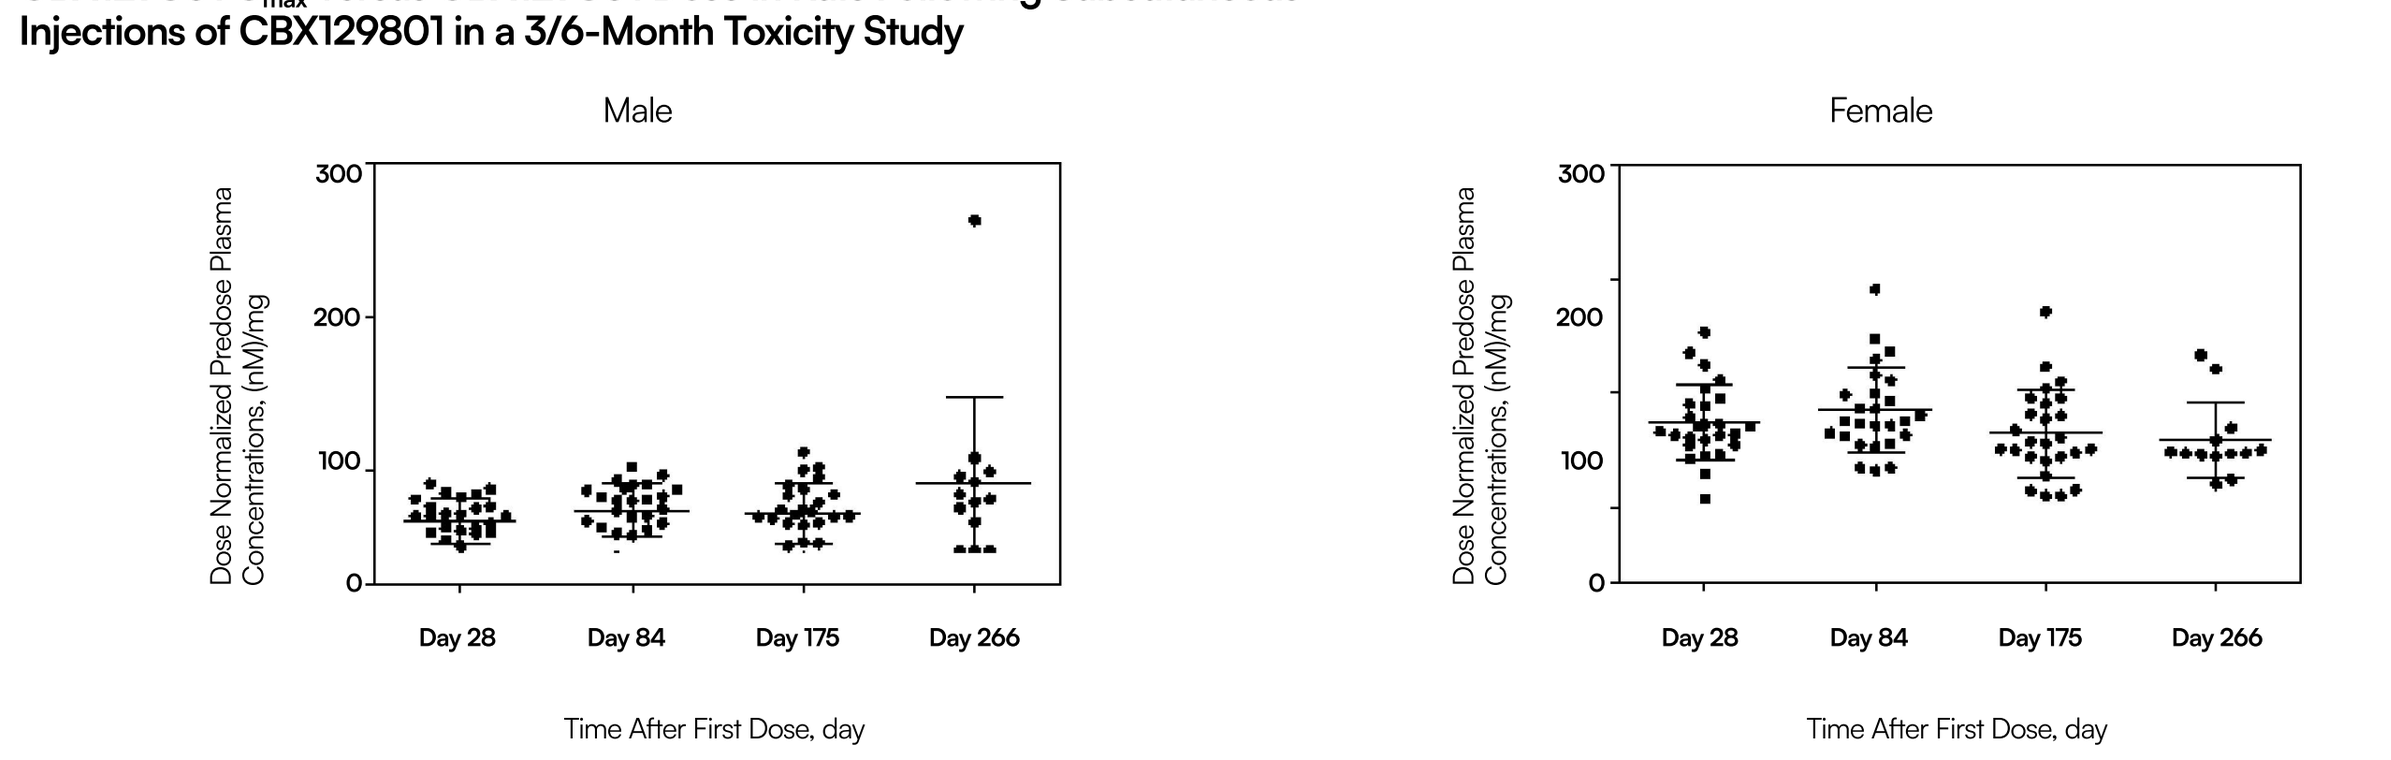

Male
Female
Dose Normalized Predose Plasma Concentrations, (nM)/mg
Day 28
Day 84
Day 175
Day 266
Time After First Dose, day

Table 1

Summary of CBX129801 Toxicokinetic Parameters in Monkeys Following Subcutaneous Injections of CBX129801 in a 3/6/9-Month Toxicity Study

TK Parameter	Male			Female			Pooled Gender		
	0.4	1.33	4.0	0.4	1.33	4.0	0.4	1.33	4.0
1st Dose (Day 0)									
C_{max}, nM	47.6	233	721	67.7	222	750	57.7	228	735
T_{max}, day[a]	1.50	2.00	1.00	1.50	2.00	1.00	1.50	2.00	1.00
$T_{1/2}$, day	2.44	3.75	3.20	2.88	3.29	3.23	2.66	3.56	3.21
AUC_{tau}, nM·day	215	1,060	3,270	308	1,070	3,410	261	1,060	3,340
AUC_{inf}, nM·day	274	1,430	4,450	427	1,720	4,700	350	1,540	4,580
C_{max} M/F Ratio	0.703	1.05	0.961	-	-	-	-	-	-
AUC_{tau} M/F Ratio	0.698	0.991	0.959	-	-	-	-	-	-
13th Dose (Day 84)									
C_{max}, nM	77.5	340	917	90.8	321	920	84.1	331	918
T_{max}, day[a]	1.50	1.50	2.00	2.00	2.00	1.00	2.00	2.00	2.00
$T_{1/2}$, day	2.38	3.18	3.05	4.17	3.62	3.77	2.83	3.35	3.61
AUC_{tau}, nM·day	357	1,600	4,460	449	1,670	4,470	403	1,640	4,460
C_{max} M/F Ratio	0.854	1.06	0.997	-	-	-	-	-	-
AUC_{tau} M/F Ratio	0.795	0.958	0.998	-	-	-	-	-	-
Day 84/0 C_{max} Ratio	1.63	1.46	1.27	1.34	1.45	1.23	1.46	1.45	1.25
Day 84/0 AUC_{tau} Ratio	1.66	1.51	1.36	1.46	1.56	1.31	1.54	1.55	1.34
26th Dose (Day 266)									
C_{max}, nM	93.3	350	1,120	98.0	364	971	95.6	357	1,050
T_{max}, day[a]	1.00	1.00	1.00	2.00	1.50	1.00	1.00	1.00	1.00
$T_{1/2}$, day	2.82	3.04	3.31	2.82	3.68	3.23	2.82	3.31	3.28
AUC_{tau}, nM·day	420	1,600	5,250	485	1,740	4,640	453	1,670	4,940
C_{max} M/F Ratio	0.952	0.962	1.15	-	-	-	-	-	-
AUC_{tau} M/F Ratio	0.866	0.920	1.13	-	-	-	-	-	-
Day 175/Day 0 C_{max} Ratio	1.96	1.50	1.55	1.45	1.64	1.29	1.66	1.57	1.43
Day 175/Day 0 AUC_{tau} Ratio	1.95	1.51	1.61	1.57	1.63	1.36	1.74	1.58	1.48
Day 175/Day 84 C_{max} Ratio	1.20	1.03	1.22	1.08	1.13	1.06	1.14	1.08	1.14
Day 175/Day 84 AUC_{tau} Ratio	1.18	1.00	1.18	1.08	1.04	1.04	1.12	1.02	1.11
39th Dose									
C_{max}, nM	80.1	295	1,030	79.1	270	815	79.6	283	924
T_{max}, day[a]	2.00	1.00	1.00	2.00	1.00	2.00	2.00	1.00	1.50
$T_{1/2}$, day	2.29	4.06	3.21	3.64	3.53	4.31	2.97	3.83	3.76
AUC_{tau}, nM·day	356	1,500	4,570	409	1,400	4,080	383	1,450	4,330
C_{max} M/F Ratio	1.01	1.09	1.26	-	-	-	-	-	-
AUC_{tau} M/F Ratio	0.870	1.07	1.12	-	-	-	-	-	-
Day 266/Day 0 C_{max} Ratio	1.68	1.27	1.43	1.17	1.22	1.09	1.38	1.24	1.26
Day 266/Day 0 AUC_{tau} Ratio	1.66	1.42	1.40	1.33	1.31	1.20	1.47	1.37	1.30
Day 266/Day 175 C_{max} Ratio	0.859	0.843	0.920	0.807	0.742	0.839	0.833	0.793	0.880
Day 266/Day 175 AUC_{tau} Ratio	0.848	0.938	0.870	0.843	0.805	0.879	0.845	0.868	0.877

ND - Not Determined

Results

CBX129801 was readily measurable in monkey plasma with median T_{max} values ranging from 1.00 to 2.00 days in both sexes (Table 1 and Figure 1).

CBX129801 mean C_{max} values were 47.6, 233 and 721 nM in males and 67.7, 222 and 750 nM in females for CBX129801 doses of 0.4, 1.33 and 4.0 mg/kg, respectively. Corresponding mean AUC_{tau} values were 215, 1060 and 3270 nM·day in males and 308, 1070 and 3410 nM·day in females. Day 1 apparent $T_{1/2}$ mean values were similar between gender and dose groups and ranged from 2.44 to 3.75 days (Table 1).

CBX129801 exposure on Day 84 following 13 once-weekly subcutaneous doses was slightly higher (~25 to 55%) than the exposure following the first dose in both sexes; however, further increases in exposure were not observed from the 13th to the 26th doses or from the 26th to the 39th doses (Tables 1 and 2 and Figures 1 through 3).

Day 266 mean C_{max} values were 80.1, 295 and 1030 nM in males and 79.1, 270 and 815 nM in females for CBX129801 doses of 0.4, 1.33 and 4.0 mg/kg/week, respectively. Corresponding mean AUC_{tau} values were 356, 1500 and 4570 nM·day in males and 409, 1400 and 4080 nM·day in females. Apparent $T_{1/2}$ mean values were similar between gender and dose groups and ranged from 2.29 to 3.31 days (Table 1).

The exposure following the first or repeated doses increased as a function of CBX129801 doses in a doseproportional manner and was similar between the genders (Table 1 and Figures 2 and 3).

CBX129801 appeared to achieve a steady state within 28 days (Figure 4) and maintained high concentrations through a week after the last dose.

Upon cessation of dosing, plasma concentrations decreased over time and were substantially low by the end of the recovery period (~ < 1 nM).

Conclusions

Upon subcutaneous administration to monkeys, CBX129801 exposure was dose proportional, had a linear TK and showed no gender-related differences. CBX129801 exposure did not increase with repeated weekly administration of 39 doses.

References

Gibaldi, M. and Perrier, D., 1982. Pharmacokinetics, Second Edition, Marcel Dekker, Inc., New York.

Acknowledgments

The Authors would like to acknowledge Teresa Morris (WIL Research Laboratories, LLC) for her oversight of the in-life portion of the TK studies.

CEBIX™