18M0300

Pharmacokinetic Bioequivalence of ATI-0918 and DOXIL®/CAELYX® in Patients with Ovarian Cancer

A. Kousba[1], N.Gallegos[2], M.Dwyer[2], G. Anderson[2];
[1]BioAgilytix, San Diego, CA, 92121. [2]Azaya Therapeutics, Inc., San Antonio, TX, 78249.

BioAgilytix

2016 AAPS
ANNUAL MEETING AND EXPOSITION
NOVEMBER 13–17, 2016
COLORADO CONVENTION CENTER, DENVER
aaps

Azaya Therapeutics

PURPOSE

Objective

To assess the pharmacokinetic (PK) bioequivalence of ATI-0918 (test product) and DOXIL®/CAELYX® (reference product).

Introduction:

Doxorubicin is an anticancer chemotherapy drug. It is an anthracycline topoisomerase inhibitor which prevents DNA replication and inhibits protein synthesis. Doxorubicin is used to treat hematological and solid cancers such as breast and ovarian carcinoma, sarcoma and many other solid tumors[1,2]. Liposomal formulations of doxorubicin are effective in reducing doxorubicin cardiotoxicity and improving its delivery to tumor sites [3,4]. DOXIL is a doxorubicin hydrochloride pegylated liposome injection which is marketed in the United States, Israel and Japan as DOXIL and elsewhere as CAELYX. DOXIL is approved by the FDA for treatment of ovarian cancer, AIDS-related Kaposi's sarcoma, and in combination therapy for multiple myeloma[5]. ATI-0918 is being developed by Azaya Inc. as a generic formulation of DOXIL/CAELYX, a liposomal formulation of doxorubicin hydrochloride.

MATERIALS

Study

This is a Single-blind, Randomized, Two-way, Crossover Bioequivalence study. Subjects with ovarian cancer were randomized in a 1:1 ratio and received one of two treatment sequences. The study enrolled 60 subjects from which 55 were evaluable for BE assessment (Five subjects were dropped out due to under-dosing).

Dose

In Sequence A, patients received a single dose of 50 mg/m² DOXIL/CAELYX on Day 1 of Cycle 1 and a single dose of 50 mg/m² ATI-0918 on Day 1 of Cycle 2. In Sequence B, patients received a single dose of 50 mg/m² ATI-0918 on Day 1 of Cycle 1 and a single dose of 50 mg/m² DOXIL/CAELYX on Day 1 of Cycle 2. There was a 3-week wash-out period between Cycle 1 and Cycle 2. Blood samples were obtained over 216 hours postdose during Cycle 1 and Cycle 2.

METHOD

Analytical Method

The plasma was analyzed for liposomal encapsulated doxorubicin, free (i.e., released) doxorubicin and doxorubicinol metabolite by a validated HPLC-MS/MS assay (Below Quantification Limit; BQL < 10.0 ng/mL for free (i.e., released) doxorubicin, 50.0 ng/mL for encapsulated doxorubicin and 0.200 ng/mL for doxorubicinol).

METHOD(CONT.)

Pharmacokinetic Analysis

The bioequivalence was performed by standard non-compartmental methods [6] based on the concentration-time data of each subject using a validated WinNonlin Professional 6.3 (Pharsight Corp., Mountain View, CA, USA). The bioequivalence of ATI-0918 (test product) to DOXIL/CAELYX (reference product) was assessed using the In-transformed $AUC_{0\text{-}t}$, $AUC_{0\text{-}inf}$, and C_{max} of the liposomal encapsulated doxorubicin, free doxorubicin, and doxorubicinol. The Least Squares Geometric Means (LSGM), %Ratio of LSGM and the lower and upper limits of the 90% Confidence Intervals (CI) of the %Ratio were calculated. Pharmacokinetic equivalence was concluded if the 90% CI for the ratio of ATI-0918 to DOXIL/CAELYX for $AUC_{0\text{-}t}$, $AUC_{0\text{-}inf}$, and C_{max} of the liposomal encapsulated doxorubicin, free doxorubicin, and doxorubicinol fall within 80.00% -125.00%.

RESULTS

Following a 50 mg/m² intravenous dose of ATI-0918 (test product) or DOXIL/CAELYX (reference product), the LSGM for the encapsulated doxorubicin C_{max} values were 41,900 and 46,900 ng/mL, $AUC_{0\text{-}t}$ were 3,650,000 and 4,220,000 ng•h/mL, and $AUC_{0\text{-}inf}$ were 4,330,000 and 4,980,000 ng•h/mL, respectively. The %Ratios of LSGM of the C_{max}, $AUC_{0\text{-}t}$ and $AUC_{0\text{-}inf}$ (ATI-0918-to-CAELYX) were 89.39%, 86.48 and 86.93%, respectively. The corresponding 90% CI for those ratios were 85.60% to 93.34%, 83.56% to 89.49% and 82.24% to 91.89%, respectively.

The LSGM for the free doxorubicin Cmax values were 3,040 and 2,970 ng/mL, $AUC_{0\text{-}t}$ were 172,000 and 179,000 ng•h/mL, and $AUC_{0\text{-}inf}$ were 220,000 and 230,000 ng•h/mL following ATI-0918 and DOXIL/CAELYX, respectively. The %Ratios of LSGM of the C_{max}, $AUC_{0\text{-}t}$ and $AUC_{0\text{-}inf}$ (ATI-0918 -to-CAELYX) were 102.48%, 95.99 and 95.66%, respectively. The corresponding 90% CI for those ratios were 90.27% to 116.34%, 90.72% to 101.57% and 88.03% to 103.95%, respectively.

The LSGM for doxorubicinol C_{max} values were 2.97 and 3.24 ng/mL and $AUC_{0\text{-}t}$ were 440 and 474 ng•h/mL following ATI-0918 and DOXIL/CAELYX, respectively. The %Ratios of LSGM of C_{max} and $AUC_{0\text{-}t}$ (ATI-0918-to- CAELYX) were 91.59 and 92.89%, respectively. The corresponding 90% CI for those ratios were 84.80% to 98.92 and 85.94% to 100.41 %, respectively. The $AUC_{0\text{-}inf}$ was incalculable due to the absence of an elimination phase.

These results indicated that the PK of free doxorubicin, encapsulated doxorubicin and the metabolite doxorubicinol following ATI-0918 was equivalent to that following DOXIL/CAELYX.

Table 1

Summary of the PK Bioequivalence for Liposomal Doxorubicin, Free Doxorubicin and Doxorubicinol in Patients with Ovarian Cancer Following a 50 mg/m2 Dose of either ATI-0918 (Test Product) or DOXIL/CAELYX (Reference Product)

PK Parameter	DOXIL/ CAELYX LSGM	ATI-0918 LSGM	%Ratio	CI 90% Lower	CI 90% Upper
Encapsulated Doxorubicin					
C_{max}	46,900	41,900	89.39	85.60	93.34
$AUC_{(0\text{-}T)}$	4,220,000	3,650,000	86.48	83.56	89.49
$AUC_{(0\text{-}inf)}$	4,980,000	4,330,000	86.93	82.24	91.89
Non-Encapsulated Doxorubicin					
C_{max}	2,970	3,040	102.48	90.27	116.34
$AUC_{(0\text{-}T)}$	179,000	172,000	95.99	90.72	101.57
$AUC_{(0\text{-}inf)}$	230,000	220,000	95.66	88.03	103.95
Doxorubicinol					
C_{max}	2,970	3,040	102.48	90.27	116.34
$AUC_{(0\text{-}T)}$	179,000	172,000	95.99	90.72	101.57

Figure 1

Mean (SD) Plasma Concentration-Time Profiles of Liposomal Doxorubicin in Patients with Ovarian Cancer Following a 50 mg/m2 Dose of either ATI-0918 (Test Product) or DOXIL/CAELYX (Reference Product)

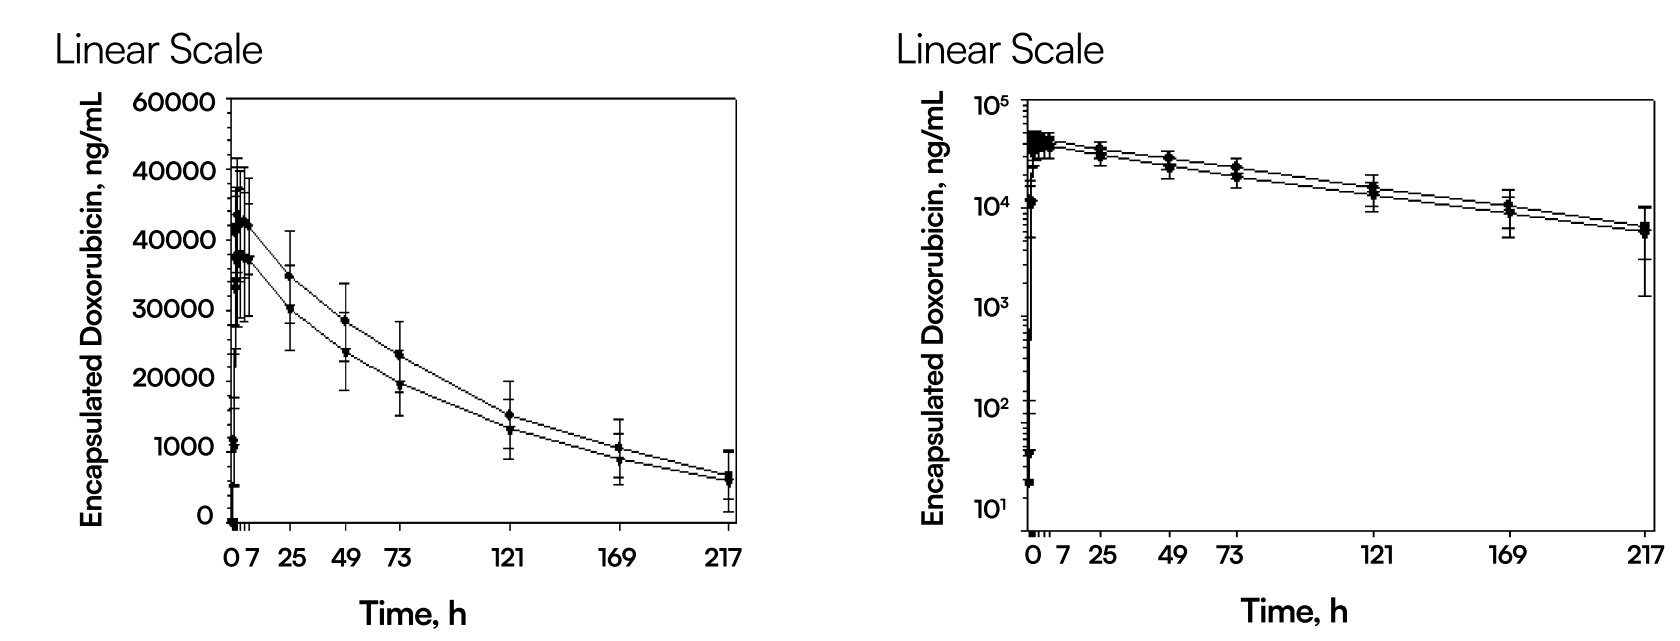
50 mg/m² Doxorubicin Dose in Caelyx
50 mg/m² Doxorubicin Dose in ATI-0918
Linear Scale
Linear Scale
Encapsulated Doxorubicin, ng/mL
Time, h

Figure 2

Mean (SD) Plasma Concentration-Time Profiles of Liposomal Doxorubicin in Patients with Ovarian Cancer Following a 50 mg/m2 Dose of either ATI-0918 (Test Product) or DOXIL/CAELYX (Reference Product)

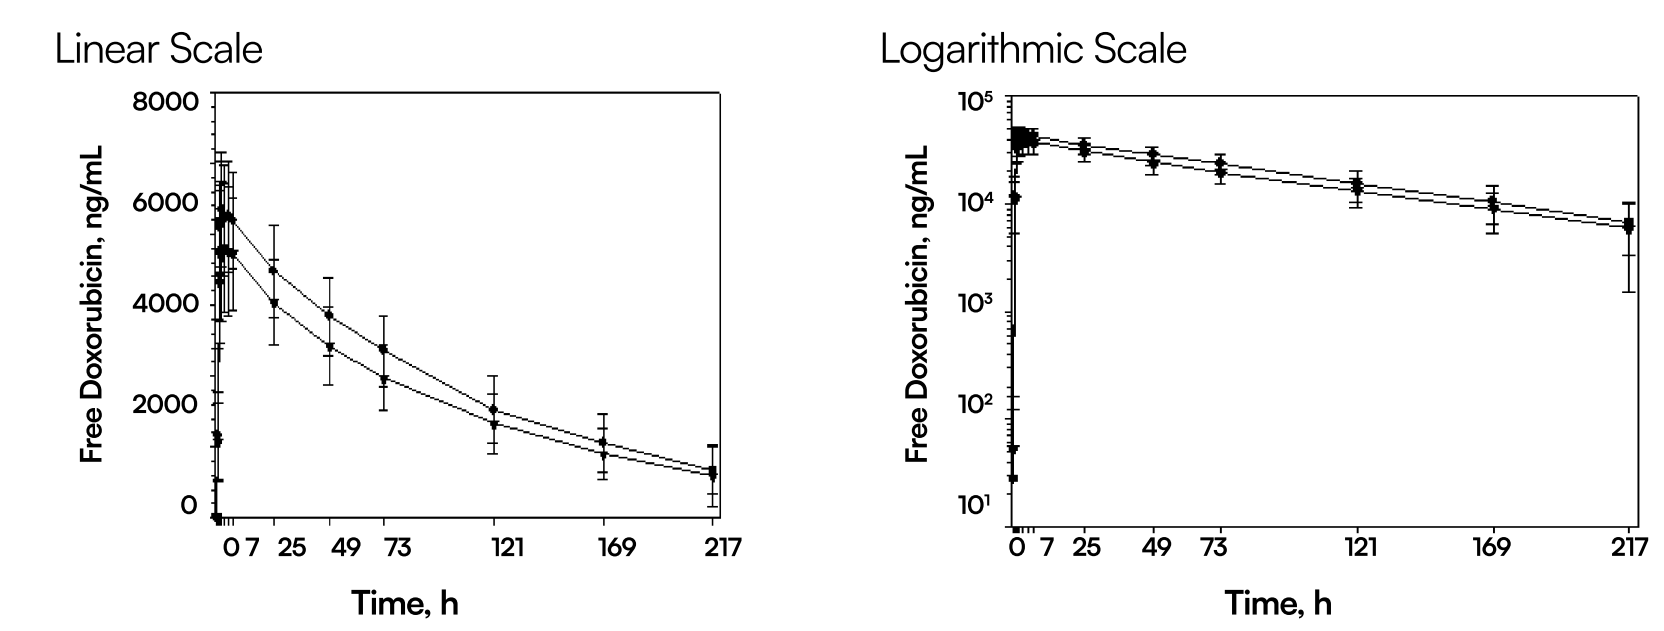
50 mg/m² Doxorubicin Dose in Caelyx
50 mg/m² Doxorubicin Dose in ATI-0918
Linear Scale
Logarithmic Scale
Free Doxorubicin, ng/mL
Time, h

Figure 3

Mean (SD) Plasma Concentration-Time Profiles of Free Doxorubicin in Patients with Ovarian Cancer Following a 50 mg/m2 Dose of either ATI-0918 (Test Product) or DOXIL/CAELYX (Reference Product)

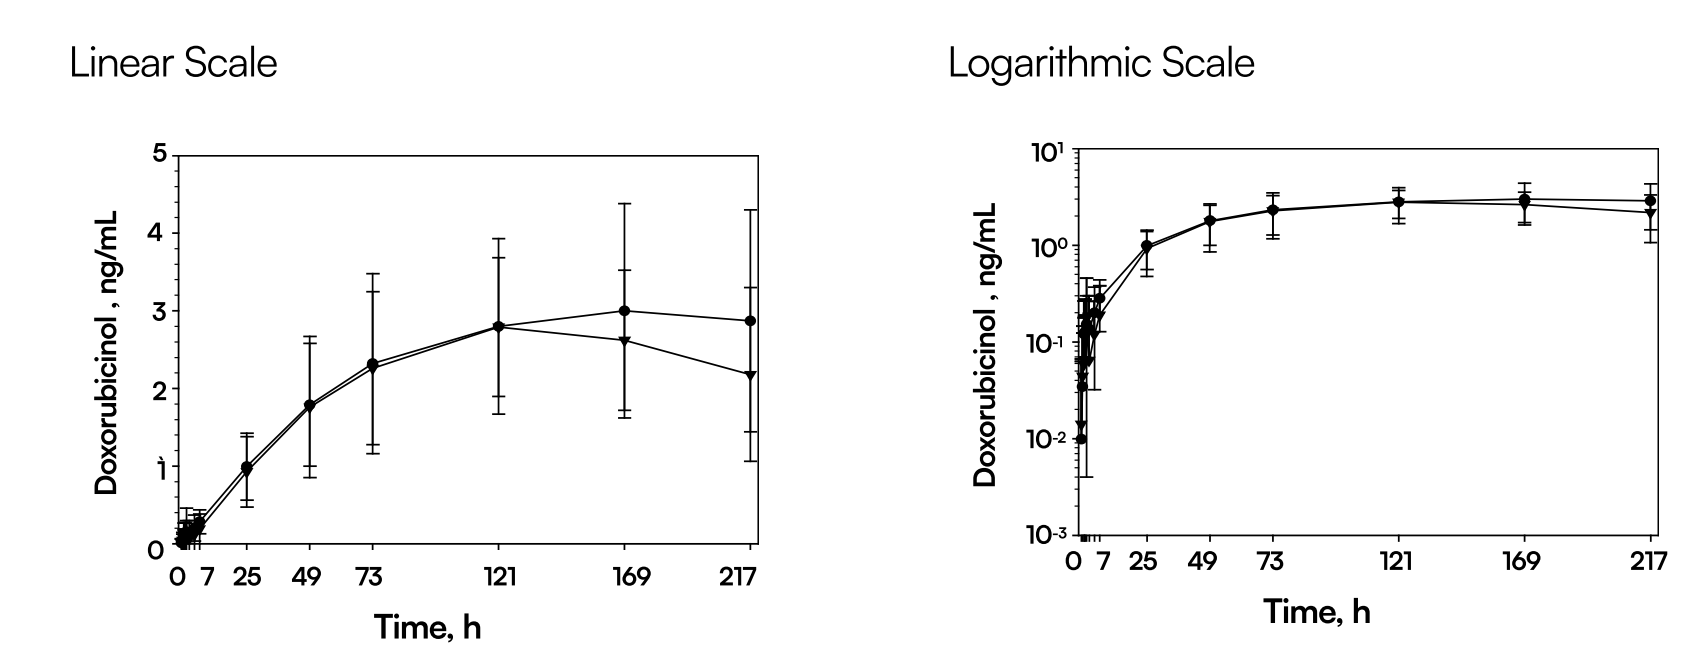
50 mg/m² Doxorubicin Dose in Caelyx
50 mg/m² Doxorubicin Dose in ATI-0918
Linear Scale
Logarithmic Scale
Doxorubicinol , ng/mL
Time, h

CONCLUSION

ATI-0918 (test product) is equivalent to DOXIL/CAELYX (reference product). The lower and higher 90% CI ratios for ln-C_{max}, ln-$AUC_{0\text{-}t}$ and ln-AUC of free doxorubicin, encapsulated doxorubicin and the metabolite doxorubicinol following ATI-0918 were within 80.00% to 125.00% of those ratios following DOXIL/CAELYX.

FUNDING / GRANTS / ENCORE / REFERENCE or other use

1. Dorr RT, Von Hoff DD. (1994). Cancer Chemotherapy Handbook, 2nd ed. Appleton and Lange, Norwalk.
2. Myers CE, Chabner BA. (1990). Anthracyclines, In: Cancer Chemotherapy; Principles and Practice, Lippincott, Philadelphia, Chabner BA, Collins JM. (eds.), pp. 356-381.
3. Gabizon A, Goren D, Cohen R, Barenholz Y. (1998). Development of liposomal anthracyclines: from basics to clinical applications. J Control Release; 53(1-3):275-279.
4. Gabizon A, Shmeeda H, Barenholz Y. (2003). Pharmacokinetics of pegylated liposomal Doxorubicin: review of animal and human studies. Clin Pharmacokinet; 42(5):419-436.
5. DOXIL® (doxorubicin HCl liposome injection) Full Prescribing Information. Centocor Ortho Biotech Products, LP, Raritan, NJ. Updated in 2010. Available at: http://www.doxil.com/assets/DOXIL_PI_Booklet. pdf.
6. Gibaldi, M. and Perrier, D. (1982). Pharmacokinetics, Second Edition, Marcel Dekker, Inc., New York.